AN OPEN-LABEL TRIAL TO ASSESS THE SAFETY OF IFX-1 IN PATIENTS WITH HIDRADENITIS SUPPURATIVA NOT ELIGIBLE FOR ADALIMUMAB

Maria Argyropoulou[1], Theodora Kanni[1], Othmar Zenker[2], Niels Riedemann[2], Evangelos J. Giamarellos-Bourboulis[1]

[1]4th Department of Internal Medicine, National and Kapodistrian University of Athens, Medical School, Greece & [2]InflaRx, Jena, Germany

INTRODUCTION & OBJECTIVES

Primary and secondary failures are often encountered in patients with moderate to severe hidradenitis suppurativa (HS) following treatment with adalimumab (ADA). This introduces as unmet need in HS. We describe a pathway of development starting from measurement of complement (C) factors in the plasma of patients and leading to the conduct of an open-label trial with one anti-C5a monoclonal antibody for patients not eligible for ADA.

COMPLEMENT CONCENTRATIONS

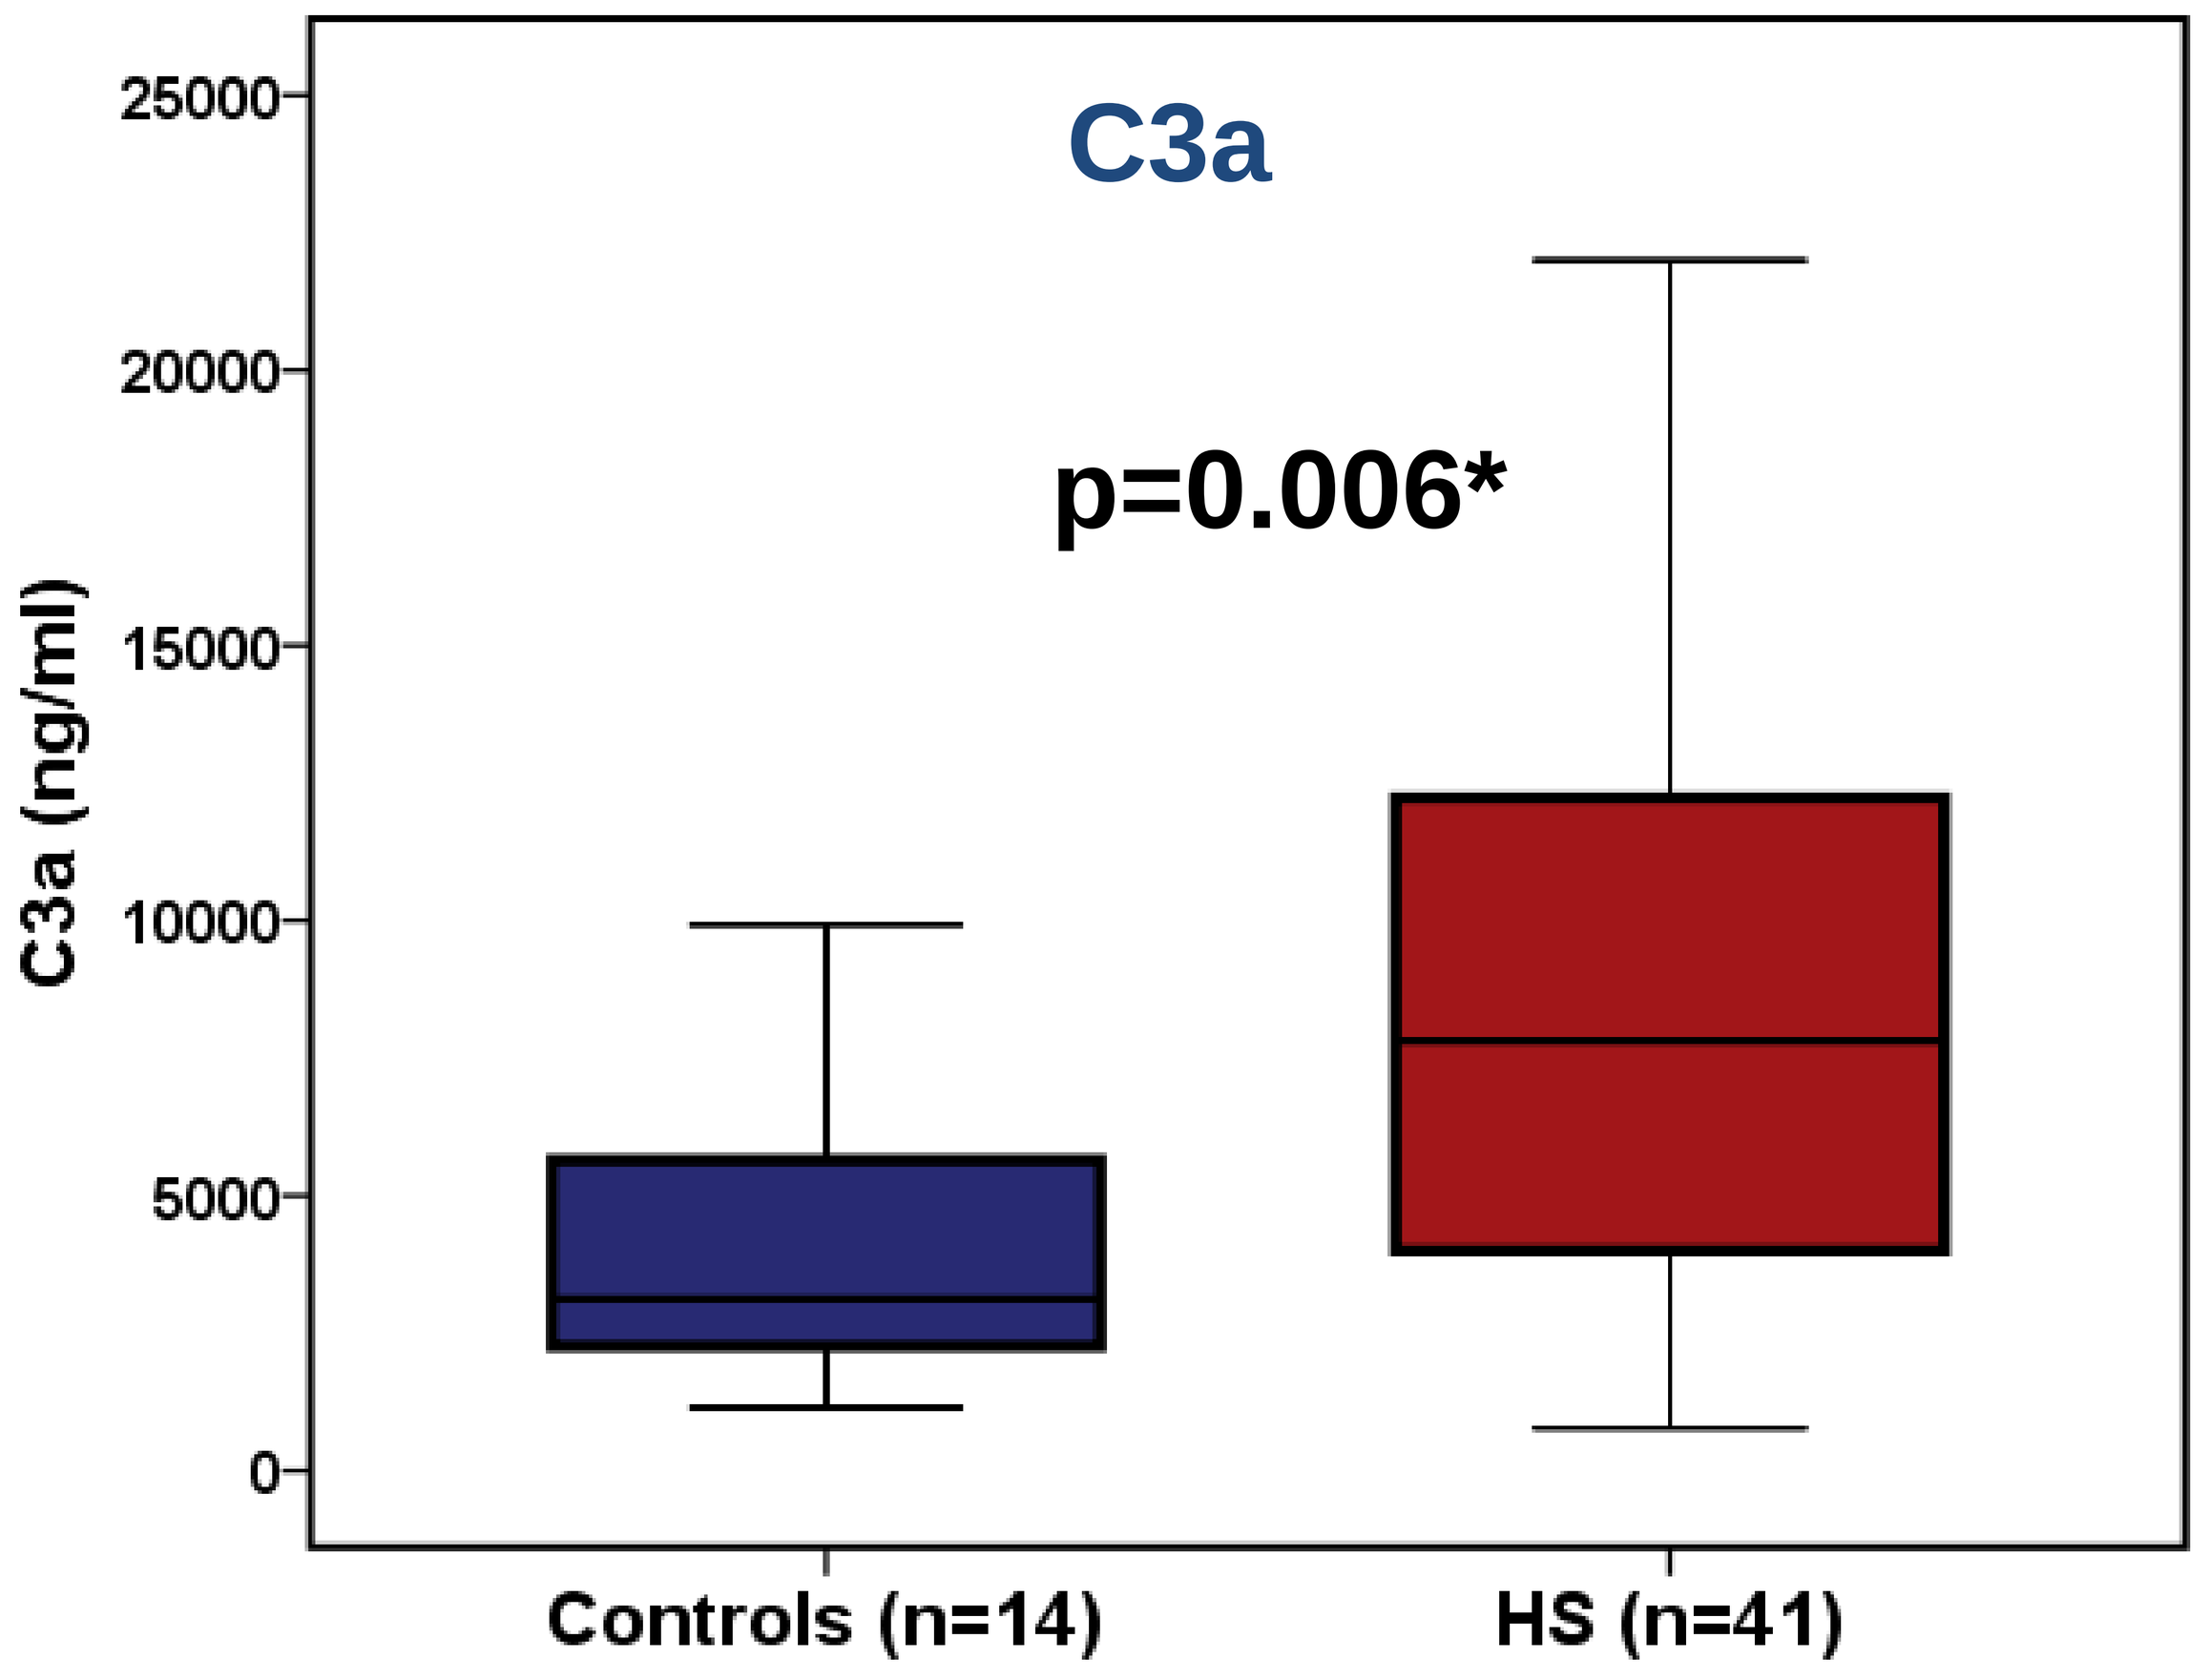
C3a
p=0.006*
C3a (ng/ml)
25000
20000
15000
10000
5000
0
Controls (n=14)
HS (n=41)

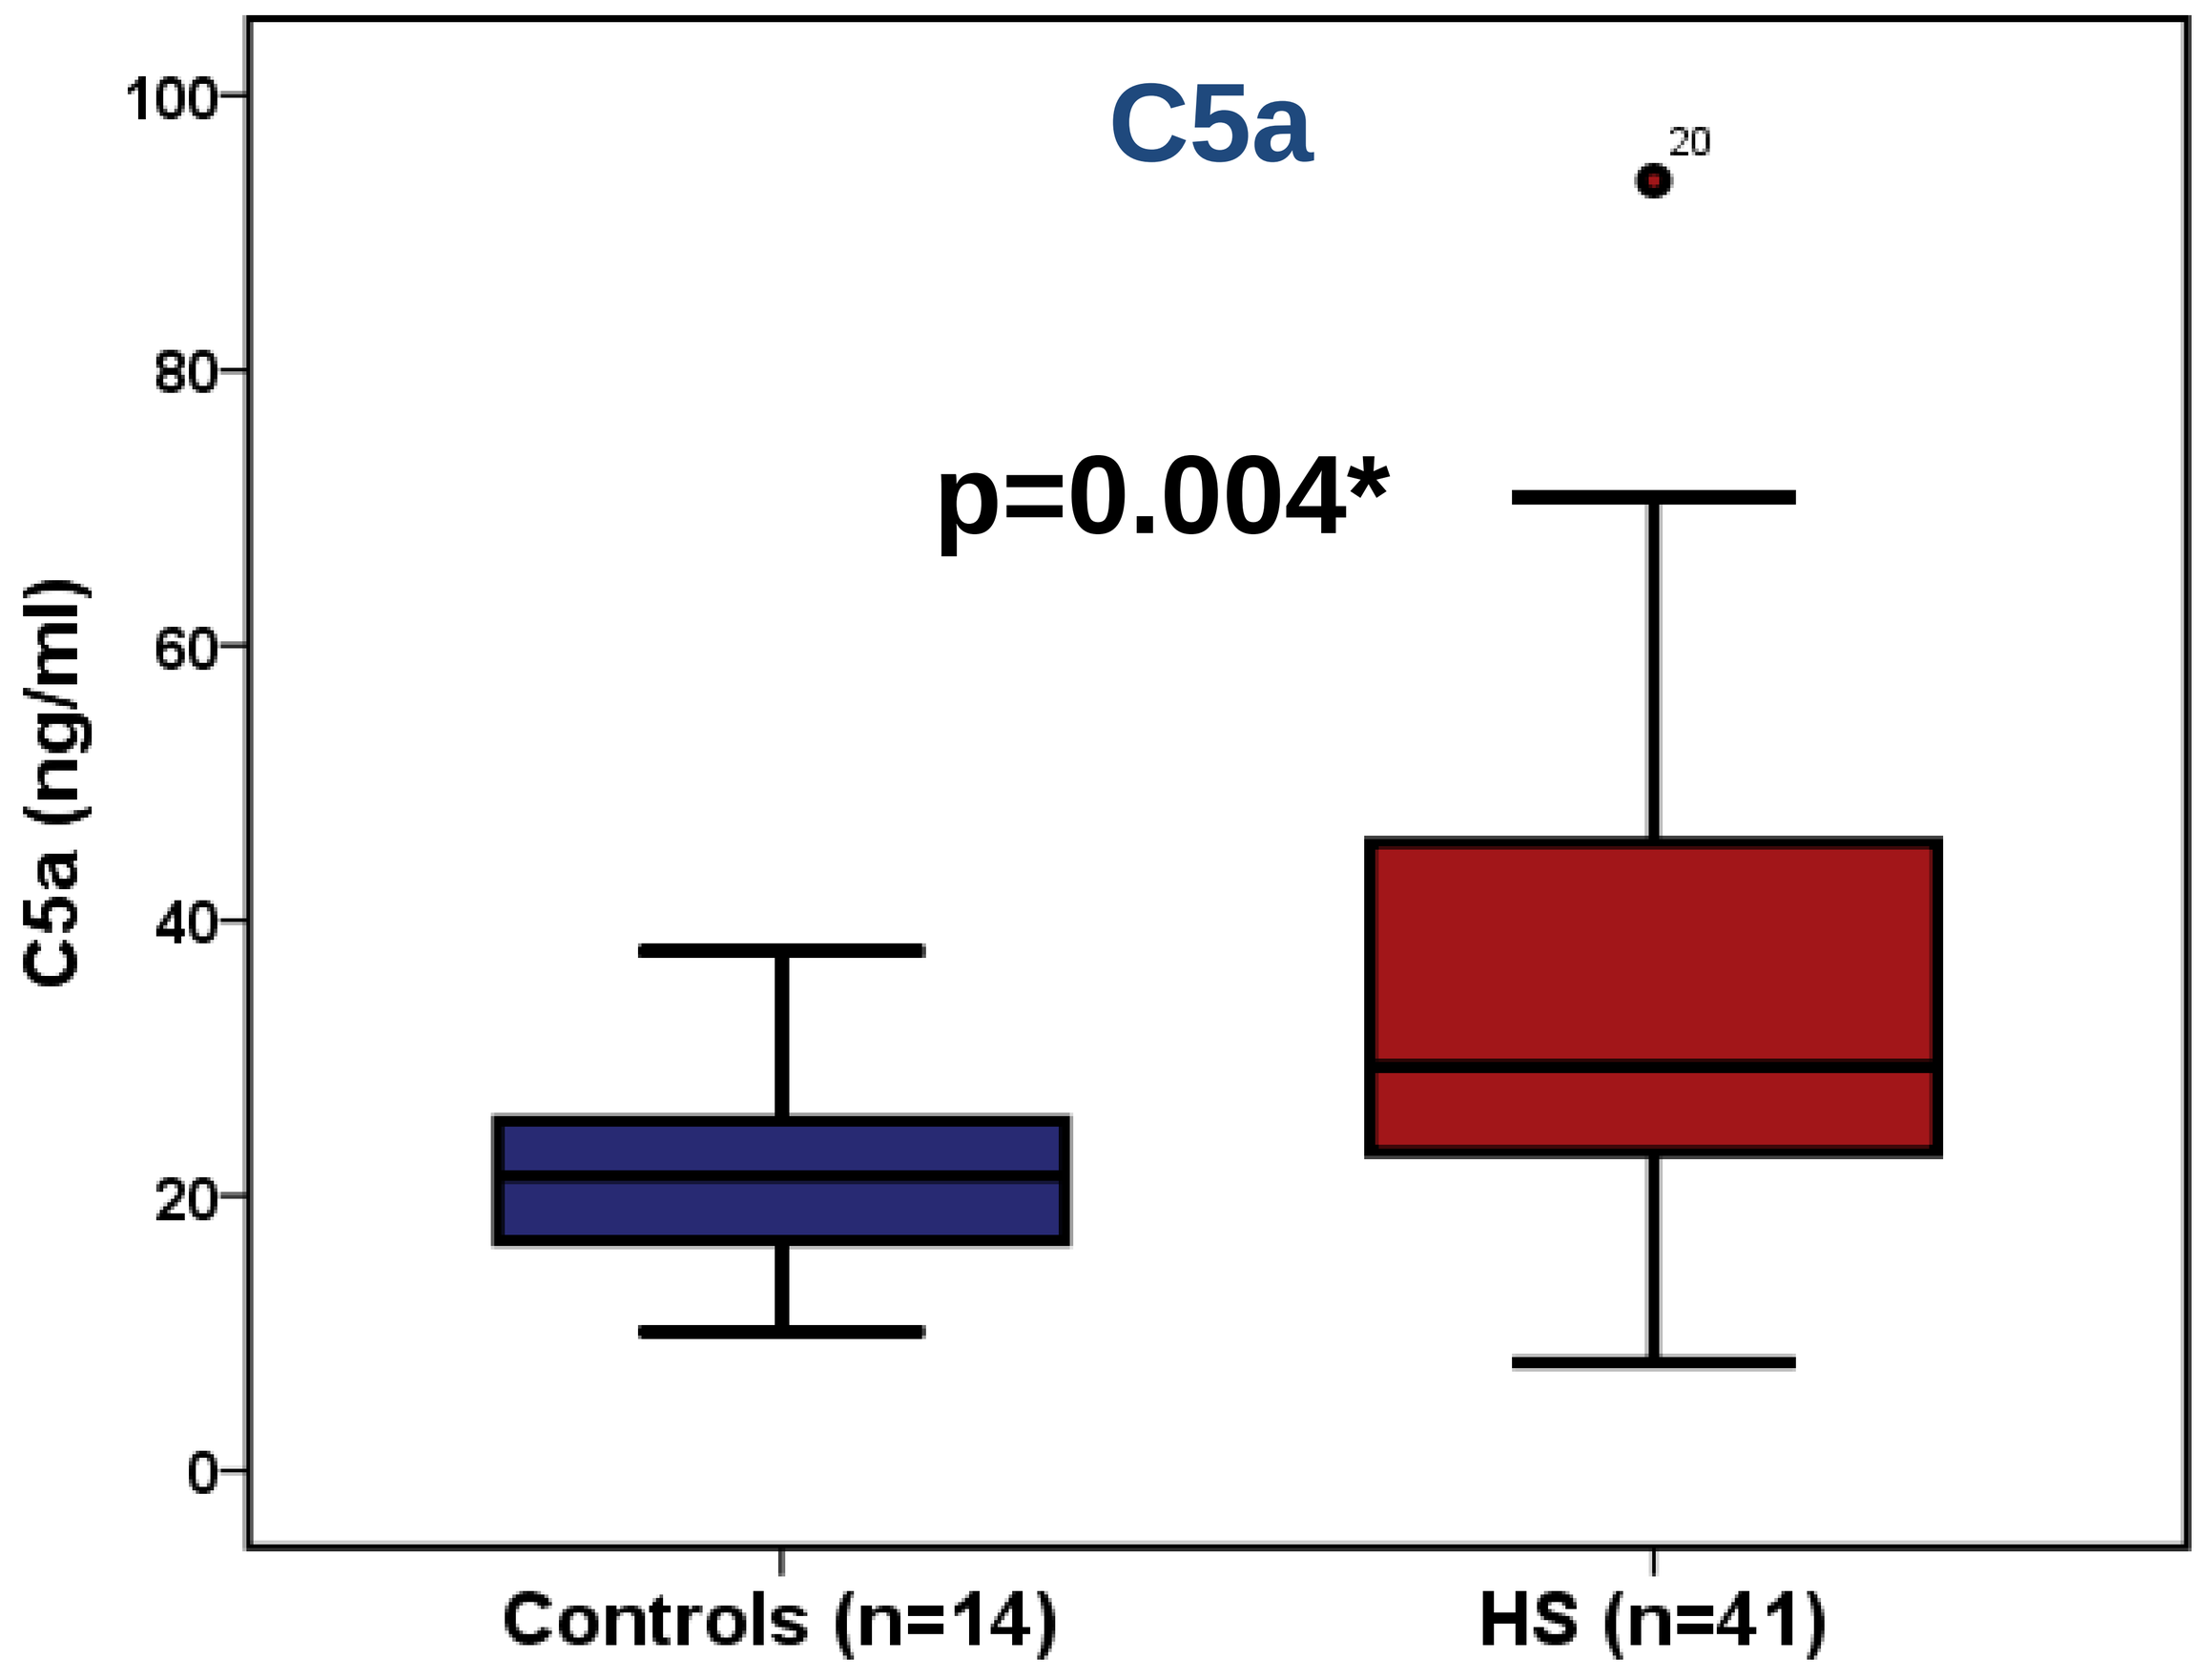
C5a
p=0.004*
C5a (ng/ml)
100
80
60
40
20
0
20
Controls (n=14)
HS (n=41)

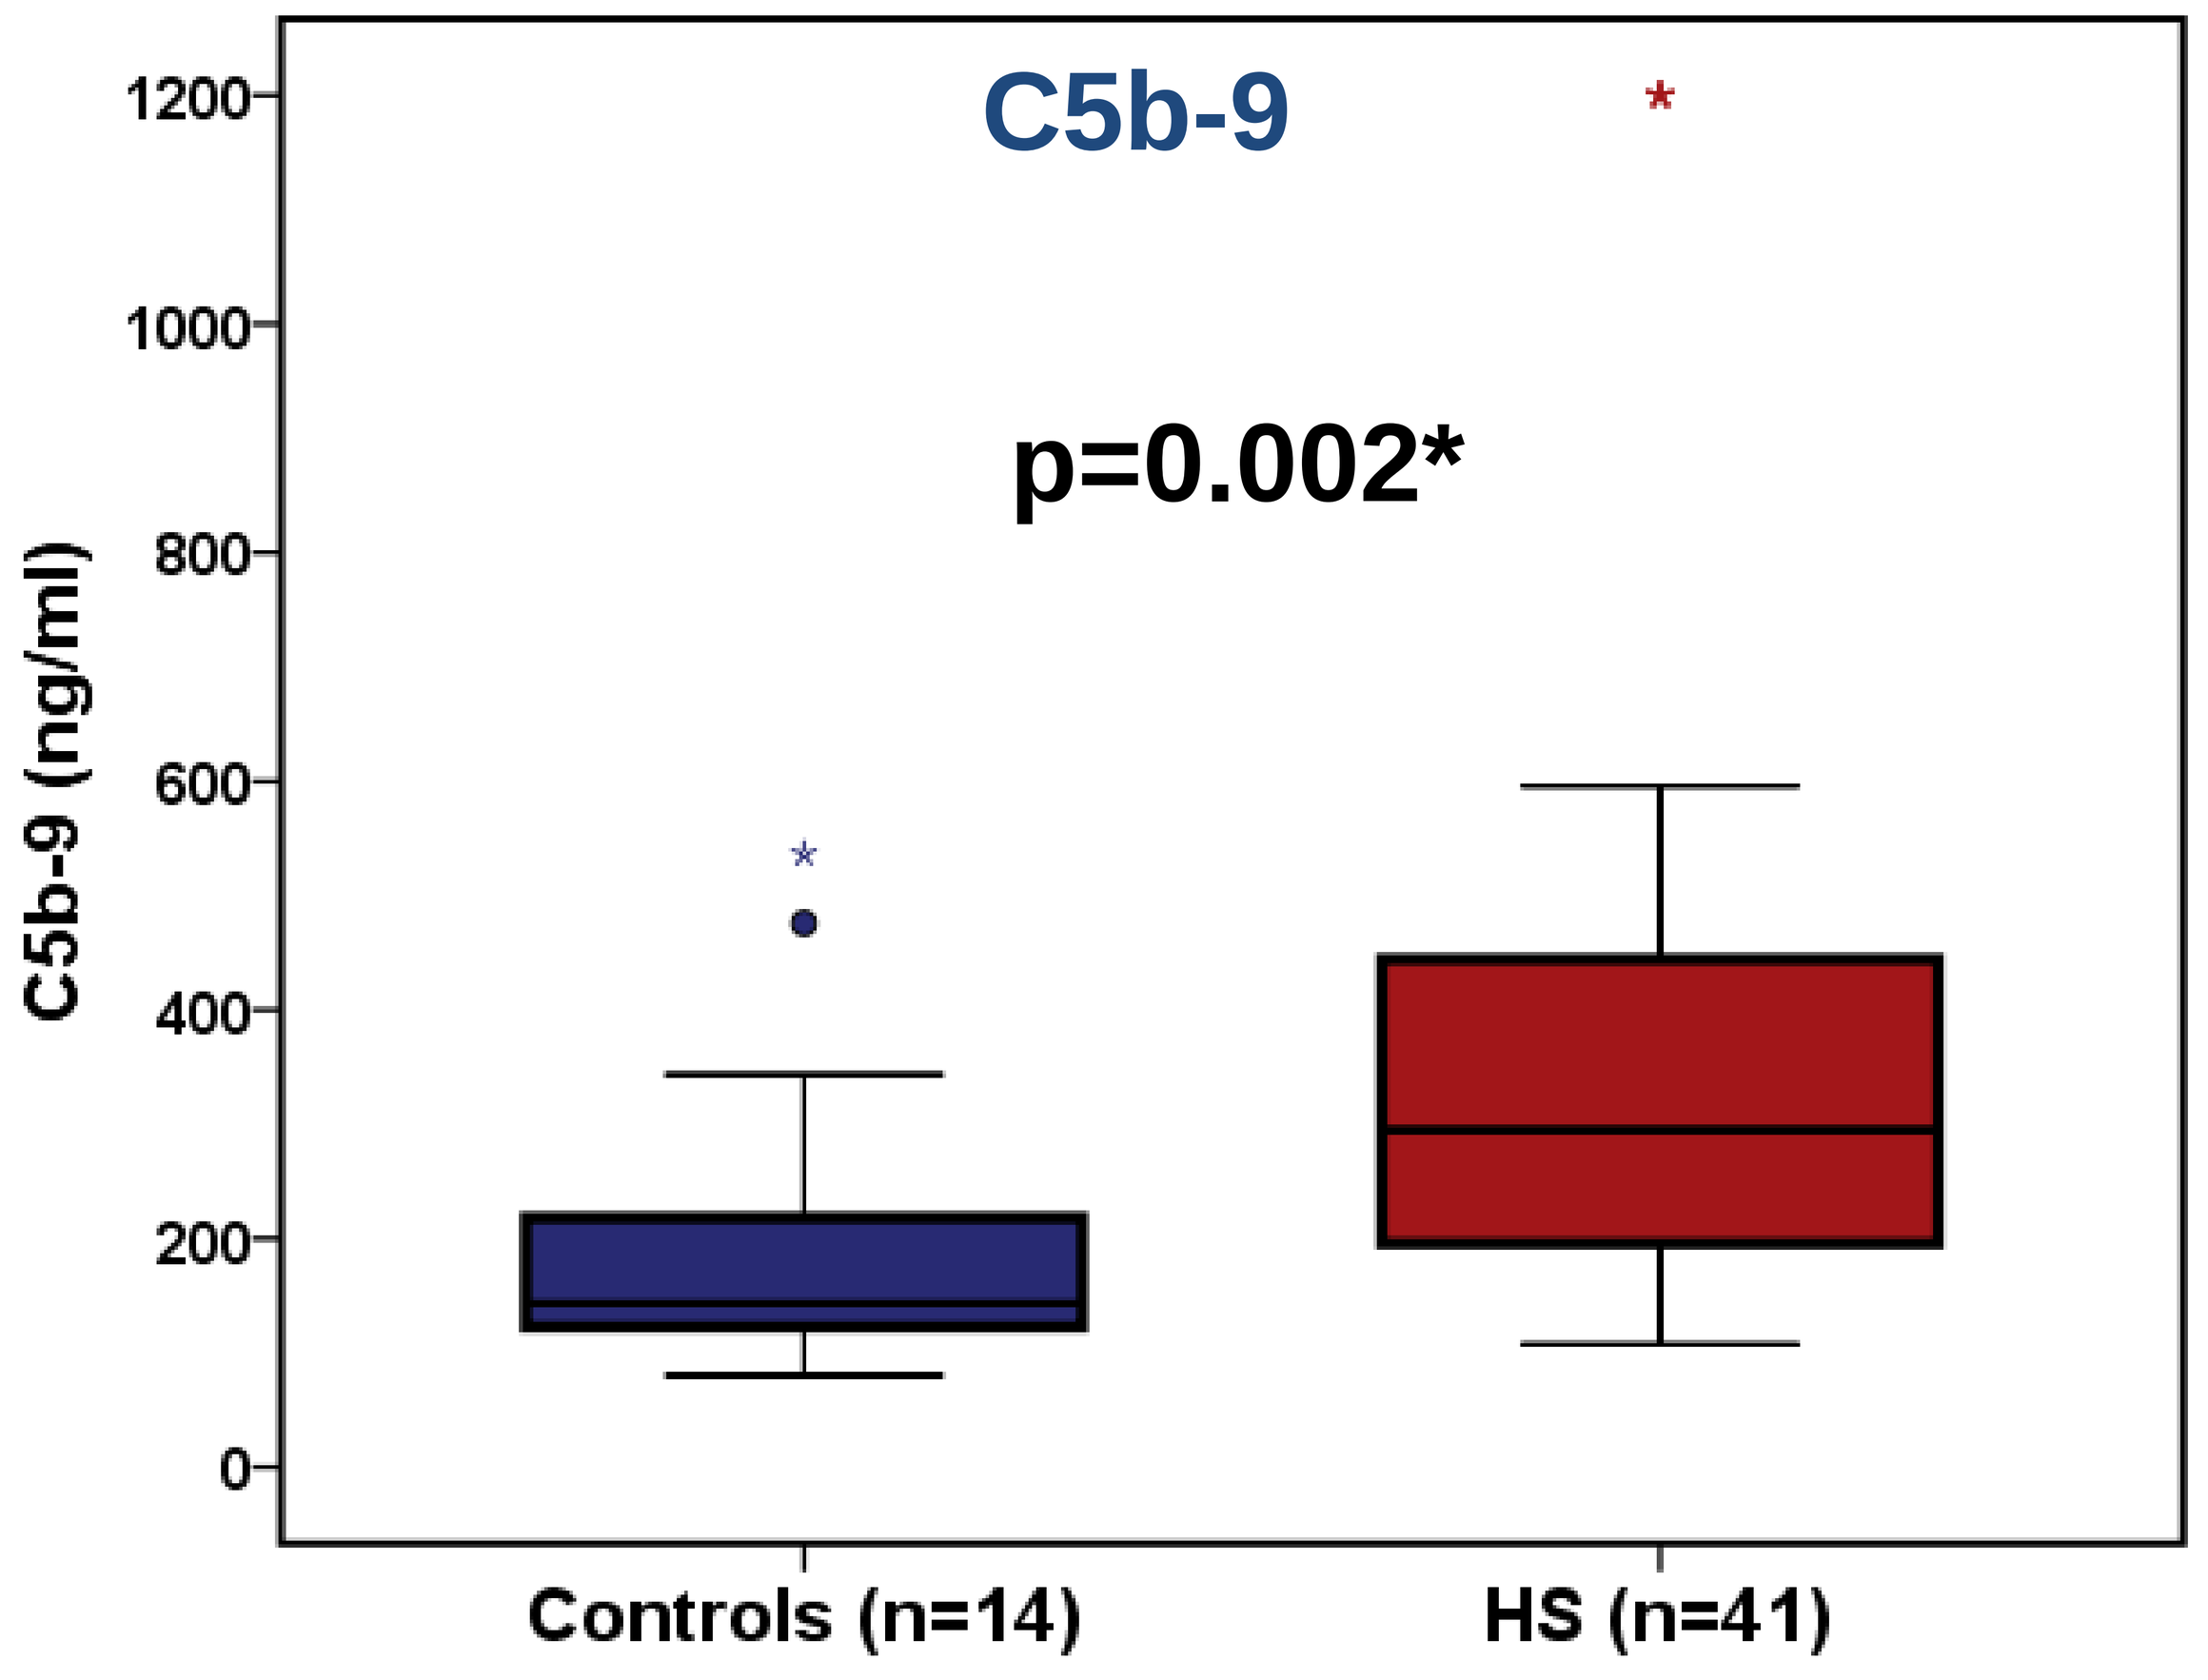
C5b-9
p=0.002*
C5b-9 (ng/ml)
1200
1000
800
600
400
200
0
Controls (n=14)
HS (n=41)

*p values of difference between controls and patients

MATERIALS & METHODS

Concentrations of C3a, C5a and of membrane attack complex (MAC) were measured in the plasma of 41 patients with moderate to severe HS and in 14 healthy individuals by an enzyme immunoassay. Based on the generated results, an open-label trial was designed and conducted. The inclusion criteria and the progress of this trial are discussed

IFX-1-P2.3 STUDY DESIGN (EudraCT 2016-002399-33)

This is an open label study in which enrolled patients are treated with IFX-1 a monoclonal antibody targeting C5a.

Inclusion criteria are:

1. Male or female patients ≥ 18 years old
2. Written informed consent
3. Diagnosis of HS for at least 1 year
4. HS lesions in at least 2 distinct anatomic areas, one of which is Hurley Stage II or III
5. Total AN (abscesses and nodules) count ≥3
6. Patients with either primary or secondary failure of biological treatment or not eligible for treatment with other biologicals
7. Failure of previous antimicrobial treatments

Major exclusion criteria are intake of corticosteroids, hepatic and renal dysfunction, active infections and co-existent rheumatologic disorders.

Intervention

All patients receive nine intravenous doses of 800 mg IFX-1 on days 1, 4, 8, 15, 22, 29, 36, 43 and 50. Safety is the primary endpoint; HS clinical response score, severity scores and skin ultrasound are the main secondary endpoints. The study is powered for 12 patients.

CONCLUSIONS & STUDY PROGRESS

- In patients with moderate to severe HS, levels of C3a, C5a and MAC are significantly elevated. This introduces the rationale that C5a inhibition may be promising novel pathway for biological therapies in HS.
- The IFX-1-P2.3 study was started in December 2016 and until end of February 2017 all 12 patients were enrolled.
- Study endpoints will be presented by the end of September 2017